HAL open science

Cyriotoxin-1a, the first toxin with antinociceptive properties purified from Cyriopagopus schioedtei spider venom: from identification to structural and functional characterization

Tânia C. Gonçalves, Evelyne Benoit, Michael Kurz, Laetitia Lucarain, Sophie Fouconnier, Stéphanie Combemale, Lucie Jaquillard, Brigitte Schombert, Rachid Boukaiba, Gerhard Hessler, et al.

To cite this version:

Tânia C. Gonçalves, Evelyne Benoit, Michael Kurz, Laetitia Lucarain, Sophie Fouconnier, et al.. Cyriotoxin-1a, the first toxin with antinociceptive properties purified from Cyriopagopus schioedtei spider venom: from identification to structural and functional characterization. Gordon Research Conference - Venom Evolution, Function and Biomedical Applications, Aug 2018, West Dover VT, United States. . cea-04466926

HAL Id: cea-04466926
https://cea.hal.science/cea-04466926

Submitted on 19 Feb 2024

HAL is a multi-disciplinary open access archive for the deposit and dissemination of scientific research documents, whether they are published or not. The documents may come from teaching and research institutions in France or abroad, or from public or private research centers.

L'archive ouverte pluridisciplinaire **HAL**, est destinée au dépôt et à la diffusion de documents scientifiques de niveau recherche, publiés ou non, émanant des établissements d'enseignement et de recherche français ou étrangers, des laboratoires publics ou privés.

Cyriotoxin-1a, the first toxin with antinociceptive properties purified from *Cyriopagopus schioedtei* spider venom: from identification to structural and functional characterization

T. Gonçalves[a,c], E. Benoit[c,d], M. Kurtz[b], L. Lucarain[a], S. Fouconnier[a], S Combemale[d], L. Jaquillard[d], B. Schombert[a], R. Boukaiba[a], G. Hessler[d], A. Bohme[a], L. Bialy[e], S. Hourcade[e], R. Béroud[f], M. De Waard[f], D. Servent[c,d], M. Partiseti[a] & **JM. Chambard[a]**

Sanofi Aventis R & D, Integrated Drug Discovery – High Content Biology, Vitry-sur-Seine, France [a] / Synthetic Molecular Design, Frankfurt, Germany [b]
Service d'Ingénierie Moléculaire des Protéines (SIMOPRO), CEA, Université Paris-Saclay, Gif sur Yvette, France [c]
Institut des Neurosciences Paris-Saclay (Neuro-PSI), UMR CNRS/Université Paris-Sud 9197, Université Paris-Saclay, Gif sur Yvette, France [d]
Sanofi R & D, Neuroscience Therapeutic Area, Neurodegeneration Research, F-91385, Chilly-Mazarin, France [e]
Smartox Biotechnology, 6 rue des Platanes, F-38120 Saint-Egrève, France [f]

$Na_V1.7$: an antinociceptive target genetically validated

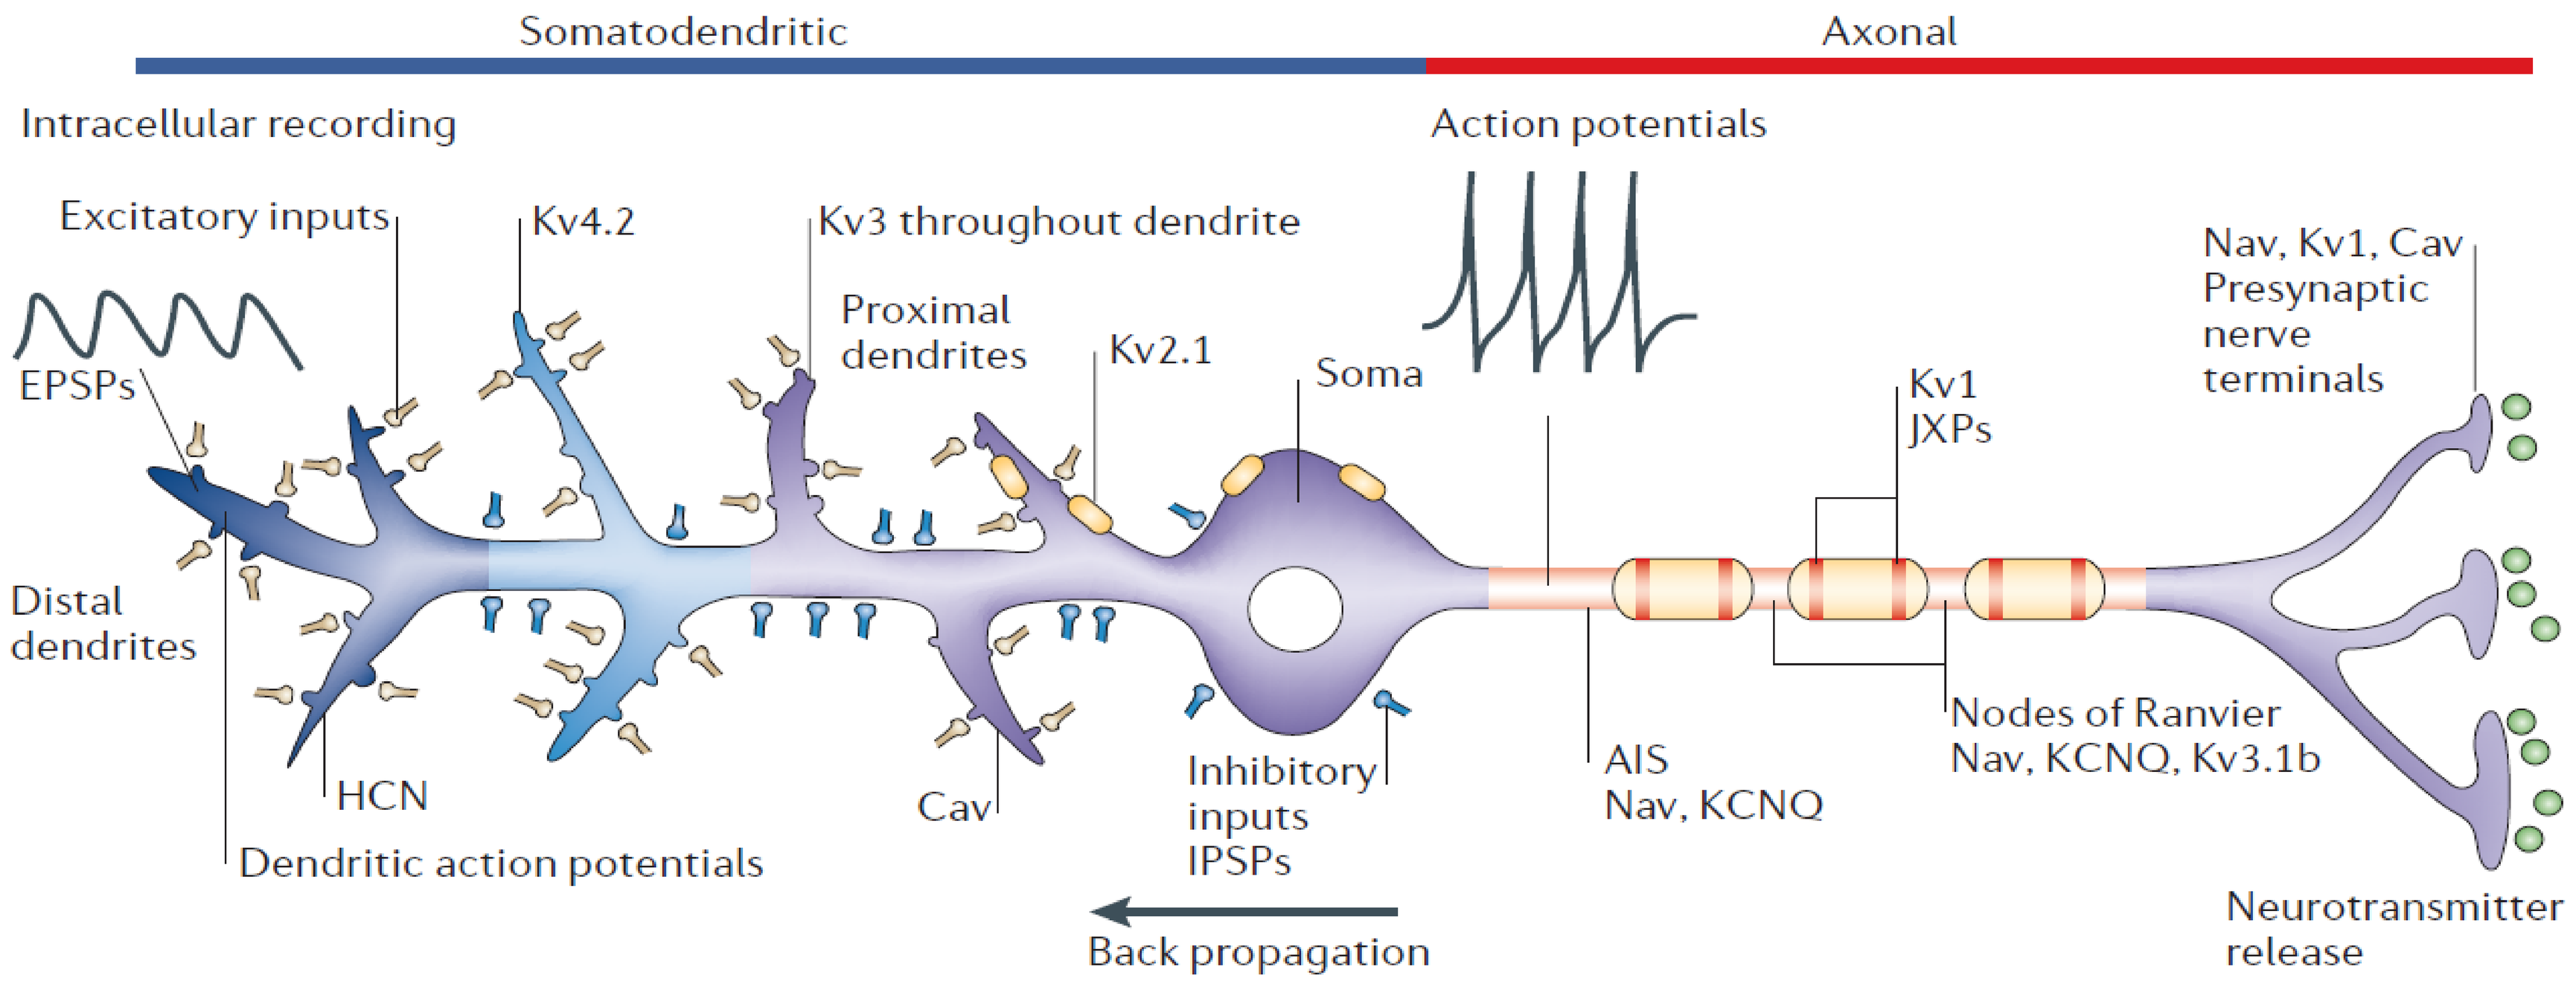
Somatodendritic
Axonal
Intracellular recording
Action potentials
Excitatory inputs
Kv4.2
Kv3 throughout dendrite
Proximal dendrites
Kv2.1
Soma
Nav, Kv1, Cav
Presynaptic nerve terminals
EPSPs
Kv1
JXPs
Distal dendrites
HCN
Cav
Inhibitory inputs
IPSPs
AIS
Nav, KCNQ
Nodes of Ranvier
Nav, KCNQ, Kv3.1b
Dendritic action potentials
Back propagation
Neurotransmitter release

$Na_V1.7$ is mainly expressed in dorsal root ganglia (DRG) neurons from peripheral to central nerve terminals (PNS). The secondary expression takes place in olfactory sensory nerves (PNS), in the pituitary gland and the subfornical organ as well as several hypothalamic nuclei (CNS) (cartoon from Lai *et al.* 2006)
Human $Na_V1.7$ genetic alterations are linked to pain :
- **$Na_V1.7$ loss of function is the root of congenital insensivity to pain (CIP) or hereditary sensory and autonomic neuropathy (HSAN) type IID.**
- **$Na_V1.7$ gain of function can cause hypersensibility to pain , primary erythromelalgia (PE), paroxysmal extreme pain disorder (PEPD), as well as small fiber neuropathy.**

In-vitro venom screening workflow

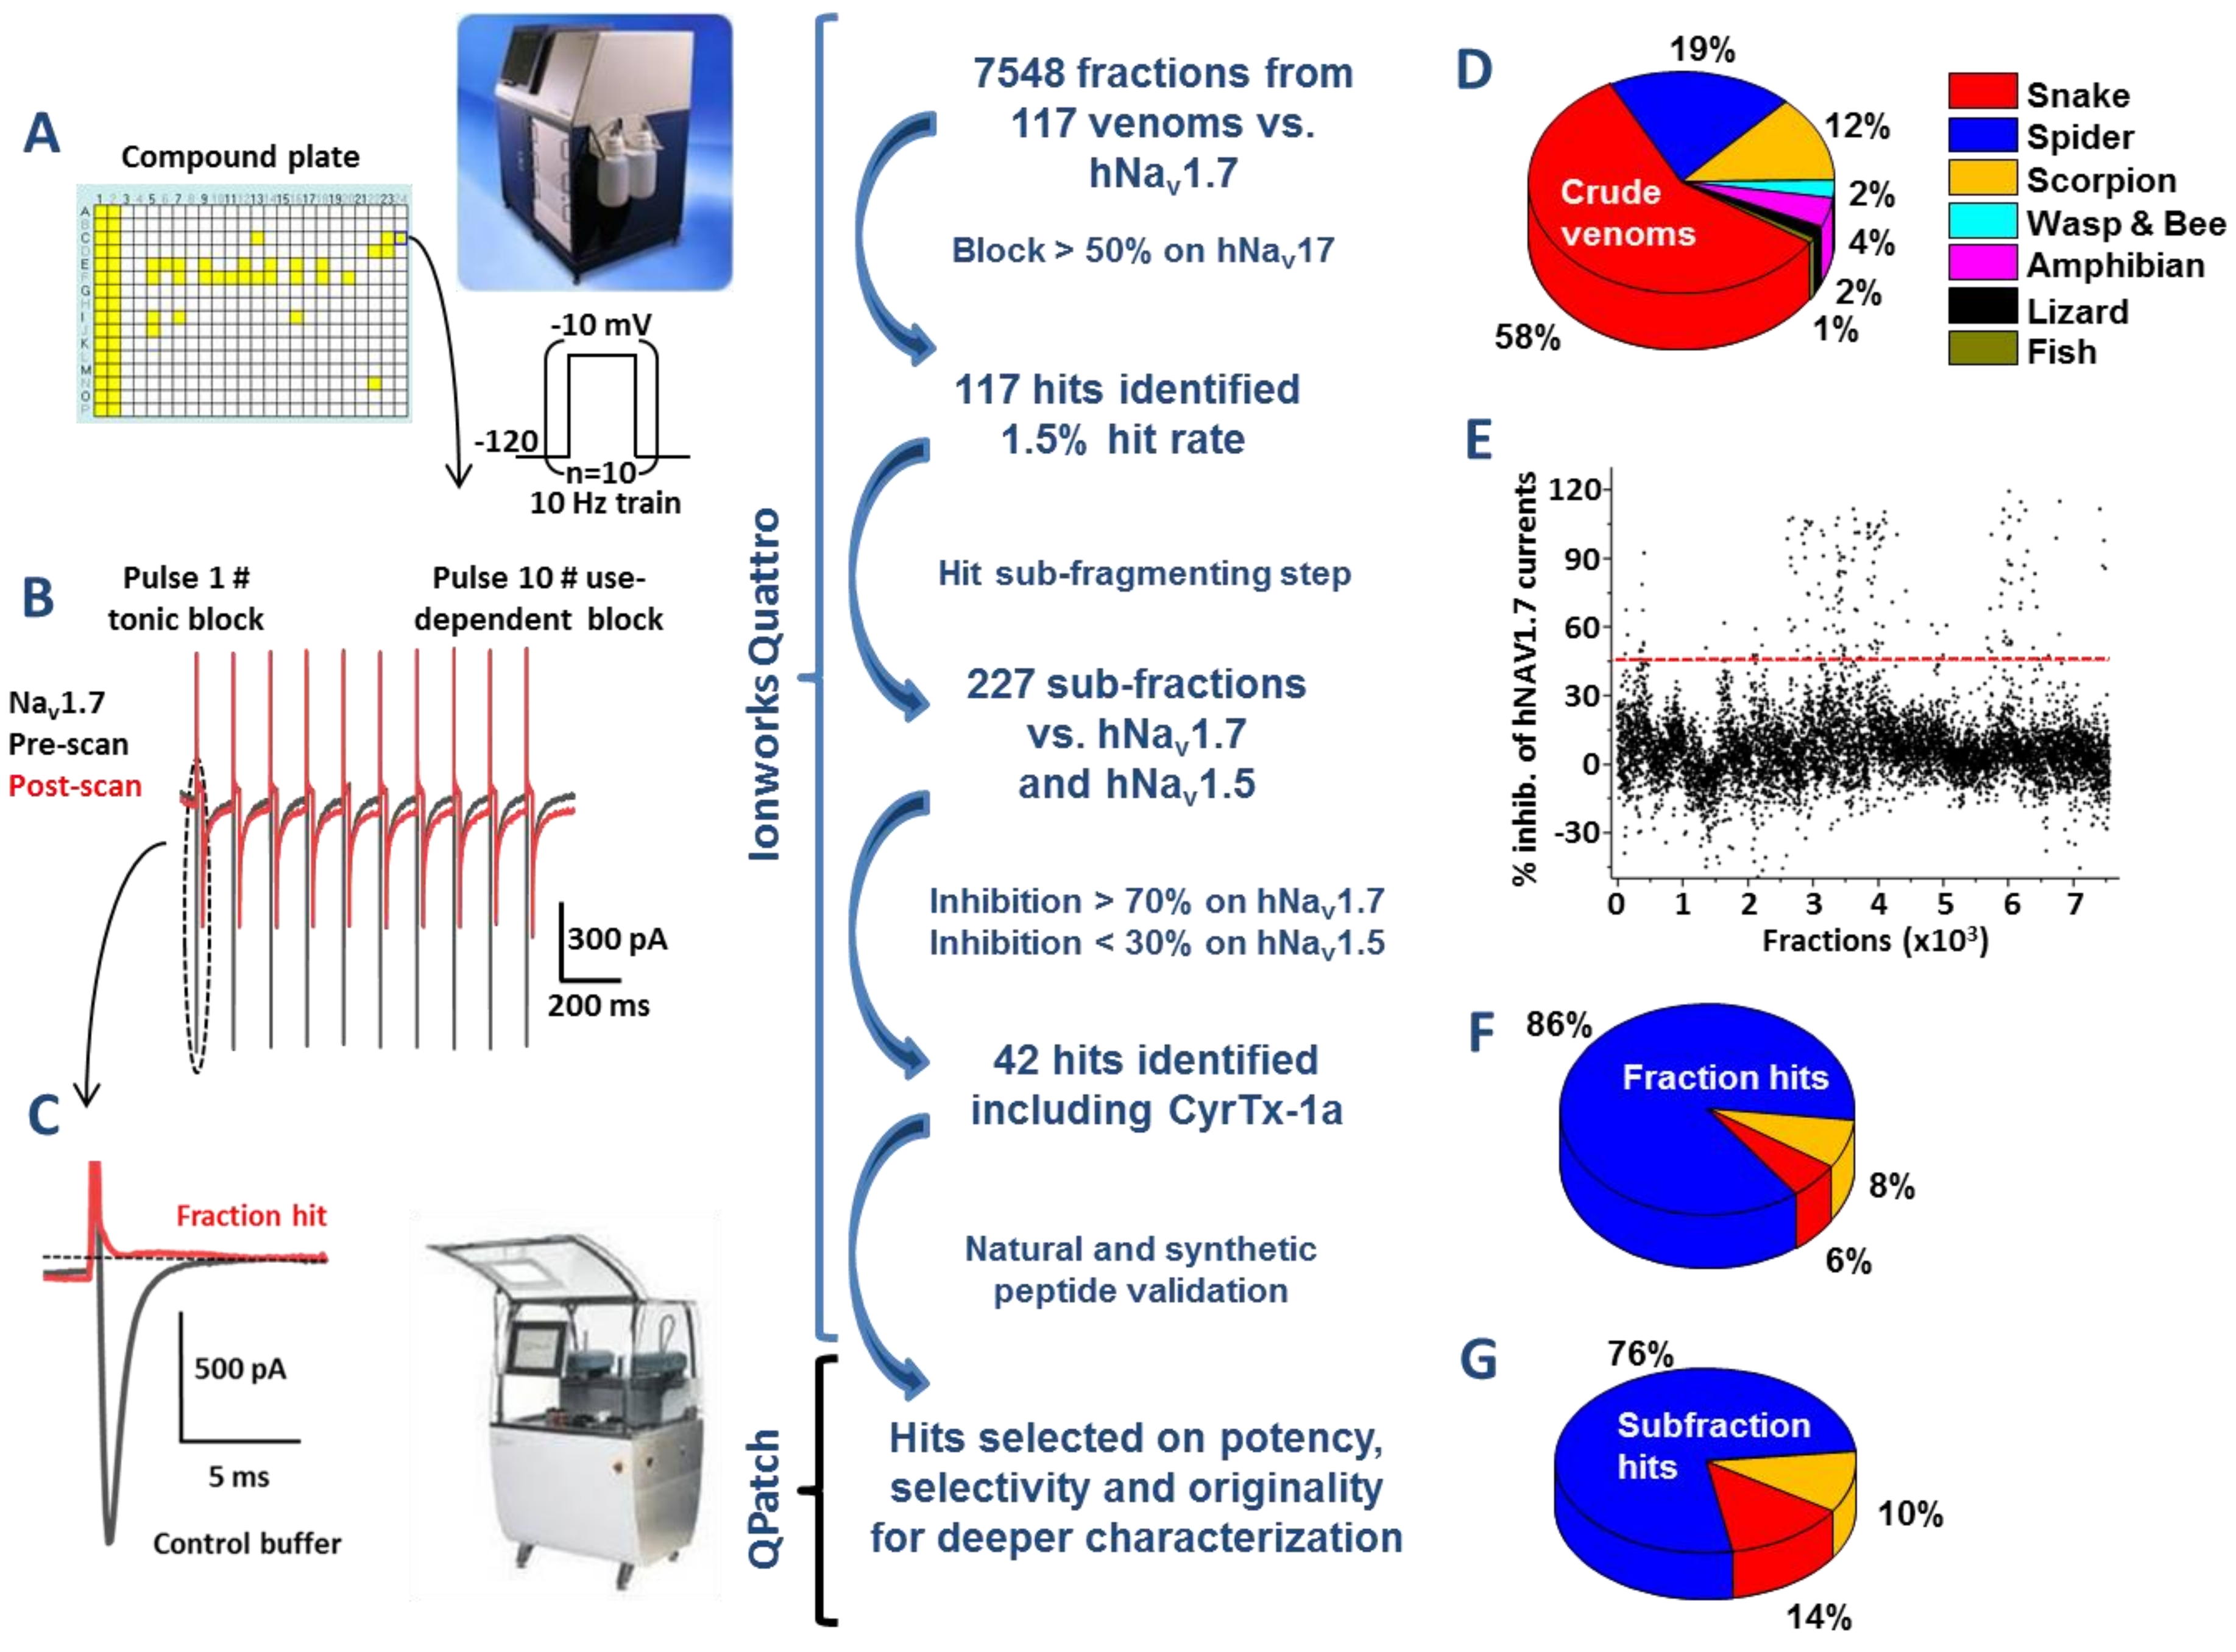
A
Compound plate
-10 mV
-120
n=10
10 Hz train
B
Pulse 1 # tonic block
Pulse 10 # use-dependent block
Na_V1.7
Pre-scan
Post-scan
300 pA
200 ms
C
Fraction hit
500 pA
5 ms
Control buffer
Ionworks Quattro
QPatch
7548 fractions from 117 venoms vs. hNaV1.7
Block > 50% on hNaV17
117 hits identified
1.5% hit rate
Hit sub-fragmenting step
227 sub-fractions vs. hNaV1.7 and hNaV1.5
Inhibition > 70% on hNaV1.7
Inhibition < 30% on hNaV1.5
42 hits identified including CyrTx-1a
Natural and synthetic peptide validation
Hits selected on potency, selectivity and originality for deeper characterization
D
Crude venoms
19%
12%
2%
4%
2%
1%
58%
Snake
Spider
Scorpion
Wasp & Bee
Amphibian
Lizard
Fish
E
% inhib. of hNAV1.7 currents
Fractions (x10³)
F
86%
Fraction hits
8%
6%
G
76%
Subfraction hits
10%
14%

(A) Collection of venoms was prepared into fractions individually added to 384-well plates (columns 1 and 2 are TTX full block and columns 3 and 4 are maximal current obtained in extracellular buffer) for testing vs. $hNa_V1.7$ on the Ionworks Quattro (IWQ, Molecular Devices). The train protocol (described as an insert) was applied before and after 10-min fraction incubation. Traces of pre-and post-scan are shown in full (B). Tonic block obtained on $hNa_V1.7$ has been enlarged (C). Source of the screened crude venom is shown in (D). Percentage of inhibition for all tested fractions was plotted. As shown in (E), a threshold was set at 50% block (red line). From the venom library tested, only 8% hits versus $hNa_V1.7$ came from scorpion venom, 6% from snake while the larger majority (86%) was spider fractions (F). (G) Hits were sub-fractioned and tested again versus $hNa_V1.7$ and $hNa_V1.5$ to identify most promising hits to be further characterized using whole-cell patch-clamp aPC (QPatch, Sophion).

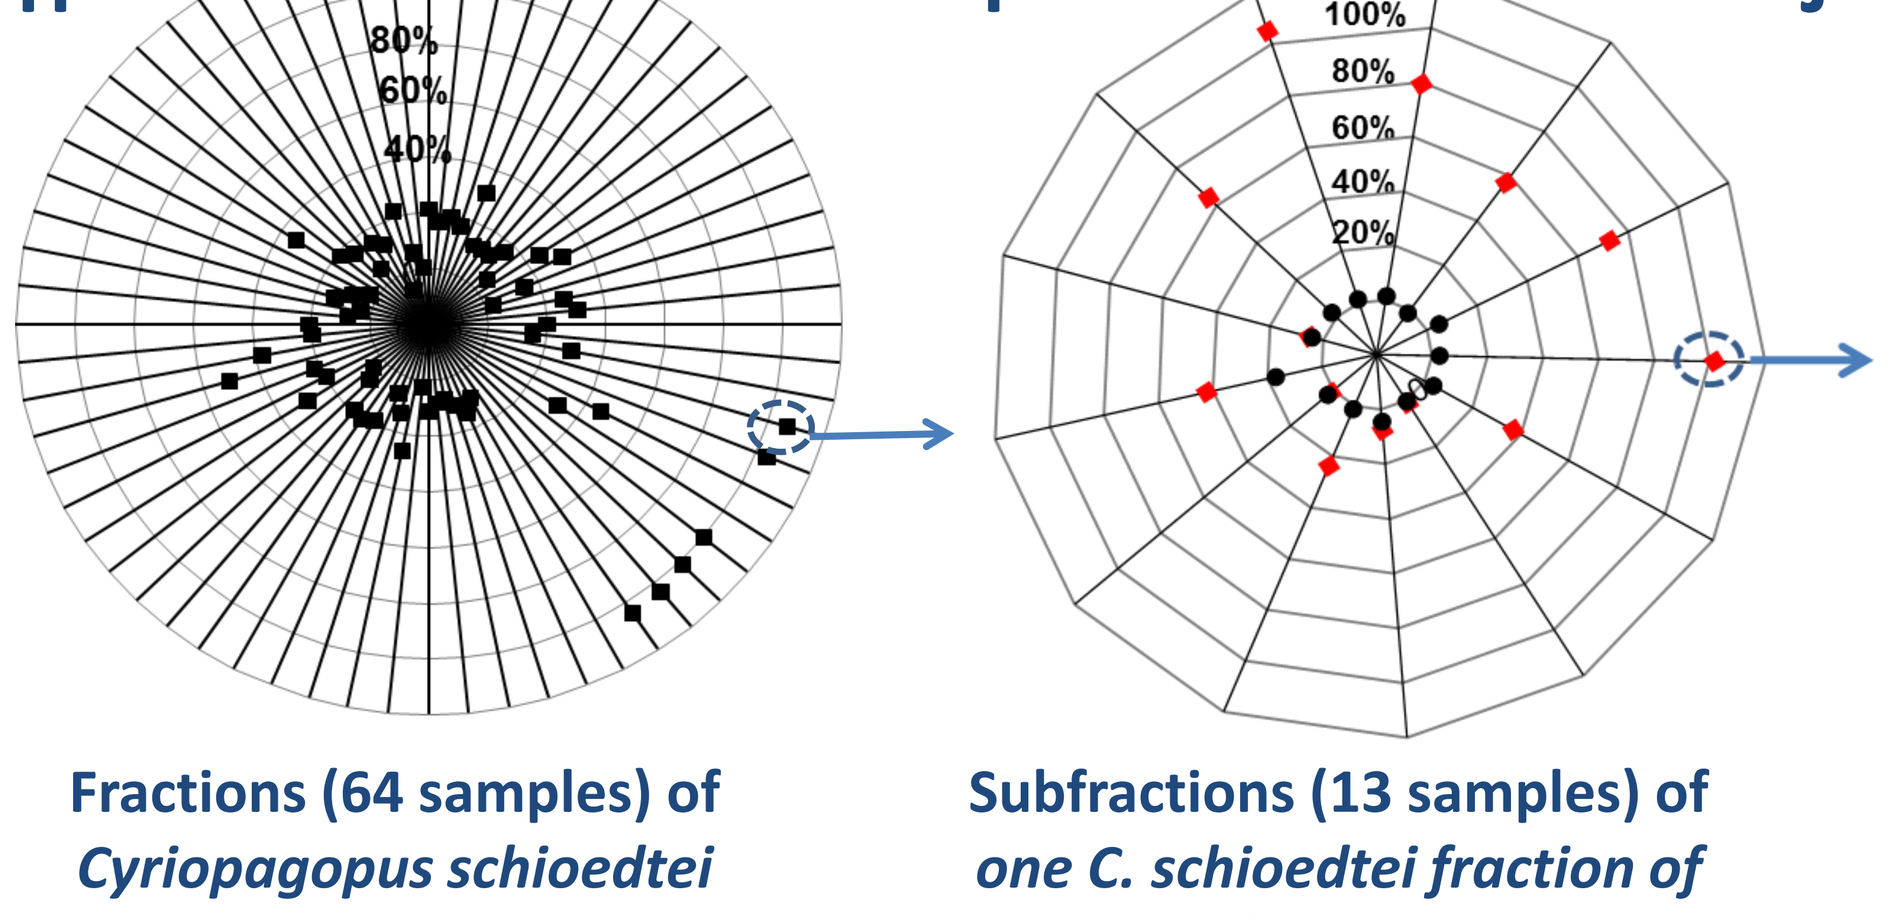
H
I
120%
100%
80%
60%
40%
20%

Fractions (64 samples) of *Cyriopagopus schioedtei* spider venom

Subfractions (13 samples) of *one C. schioedtei fraction of interest*

Flowchart for $hNa_V1.7$ hit peptide identification. (H) Percentage of inhibition of peak $hNa_V1.7$ elicited current by application of 0.05 µg of each of the 64 fractions obtained from *C. schioedtei* spider venom. (I) Each fraction was then separated again to isolate one peptide per well. Sub-fractions were tested again in our IWQ assay. At this stage, $hNa_V1.7$ (red squares) and $hNa_V1.5$ (black circles) inhibition was investigated.

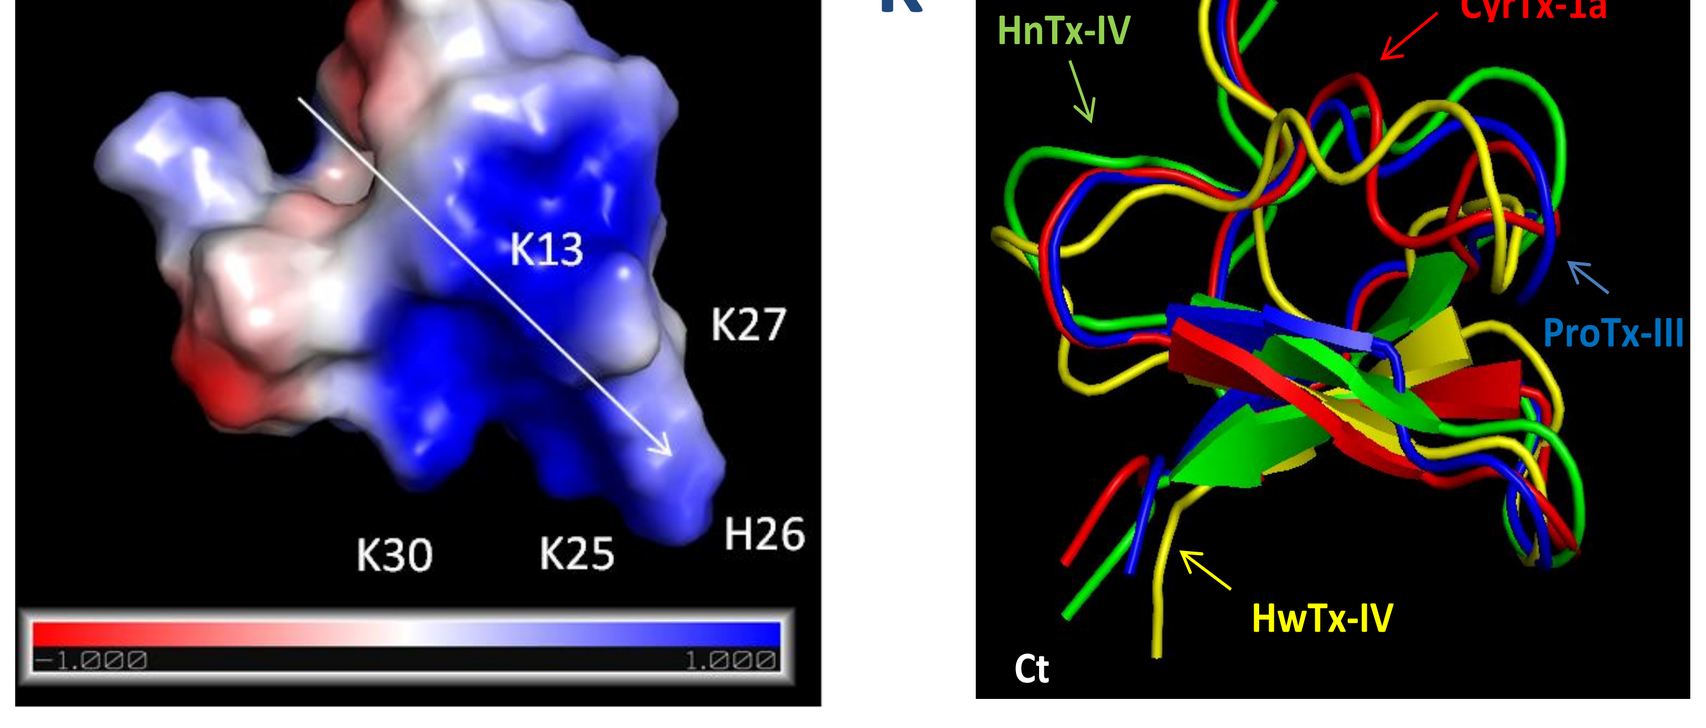
J
E15
K13
K27
K30
K25
H26
K
Nt
HnTx-IV
CyrTx-1a
ProTx-III
HwTx-IV
Ct

Representation of CyrTx-1a identified by PyMOL. (J) Electrostatic charged surface representation of CyrTx-1a. The molecule is rendered as a surface colored according to the electrostatic potential. The entire structure has a clear dipole potential with E1 and E15 forming a negative zone while K3, K7, K13, K25, H26, K27 and K30 form a positive zone. (K) Superposition of backbone peptide folding of CyrTx-1a and three other toxins of the NaSpTx family 1 previously described to possess analgesic effects (PDB entries of HnTx-IV: 1NIY, ProTx-III: 2MXM and HwTx-IV: 1MB6).

Inhibitory toxins : potential painkillers targeting $Na_V1.7$

- ProTx-II — $IC_{50}(hNa_V1.7)$ = 0.3 nM
- JzTx-V — IC_{50} $(hNa_V1.7)$ = 0.6 nM
- Pn3a — IC_{50} $(hNa_V1.7)$ = 0.9 nM
- Df1a — IC_{50} $(hNa_V1.7)$ = 1.9 nM
- CcoTx-1 — IC_{50} $(hNa_V1.7)$ = 2.0 nM
- JzTx-34 — IC_{50} $(hNa_V1.7)$ = 610 nM
- ProTx-III — IC_{50} $(hNa_V1.7)$ = 2.5 nM
- Phlo 1a, 1b, 2a — IC_{50} $(hNa_V1.7)$ ≈ 400 nM
- BDS-1 — IC_{50} $(hNa_V1.7)$ = 3.0 nM
- GrTx-1 — IC_{50} $(hNa_V1.7)$ = 370 nM
- GpTx-1 — IC_{50} $(hNa_V1.7)$ = 4.4 nM
- HnTx-III — IC_{50} $(hNa_V1.7)$ = 232 nM
- Cd1a — IC_{50} $(hNa_V1.7)$ = 16.0 nM
- Hd1a — $ICv(hNa_V1.7)$ = 110 nM
- HwTx-IV — IC_{50} $(hNa_V1.7)$ = 17.0 nM
- **CyrTx-1a — $IC_{50}(hNa_V1.7)$ = 59.0 nM**
- ProTx-I — $ICv(hNa_V1.7)$ = 51.0 nM
- GsAFI — IC_{50} $(hNa_V1.7)$ = 40.0 nM
- Ssm6a — IC_{50} $(hNa_V1.7)$ = 25.0 nM
- HnTx-IV — IC_{50} $(hNa_V1.7)$ = 21.0 nM

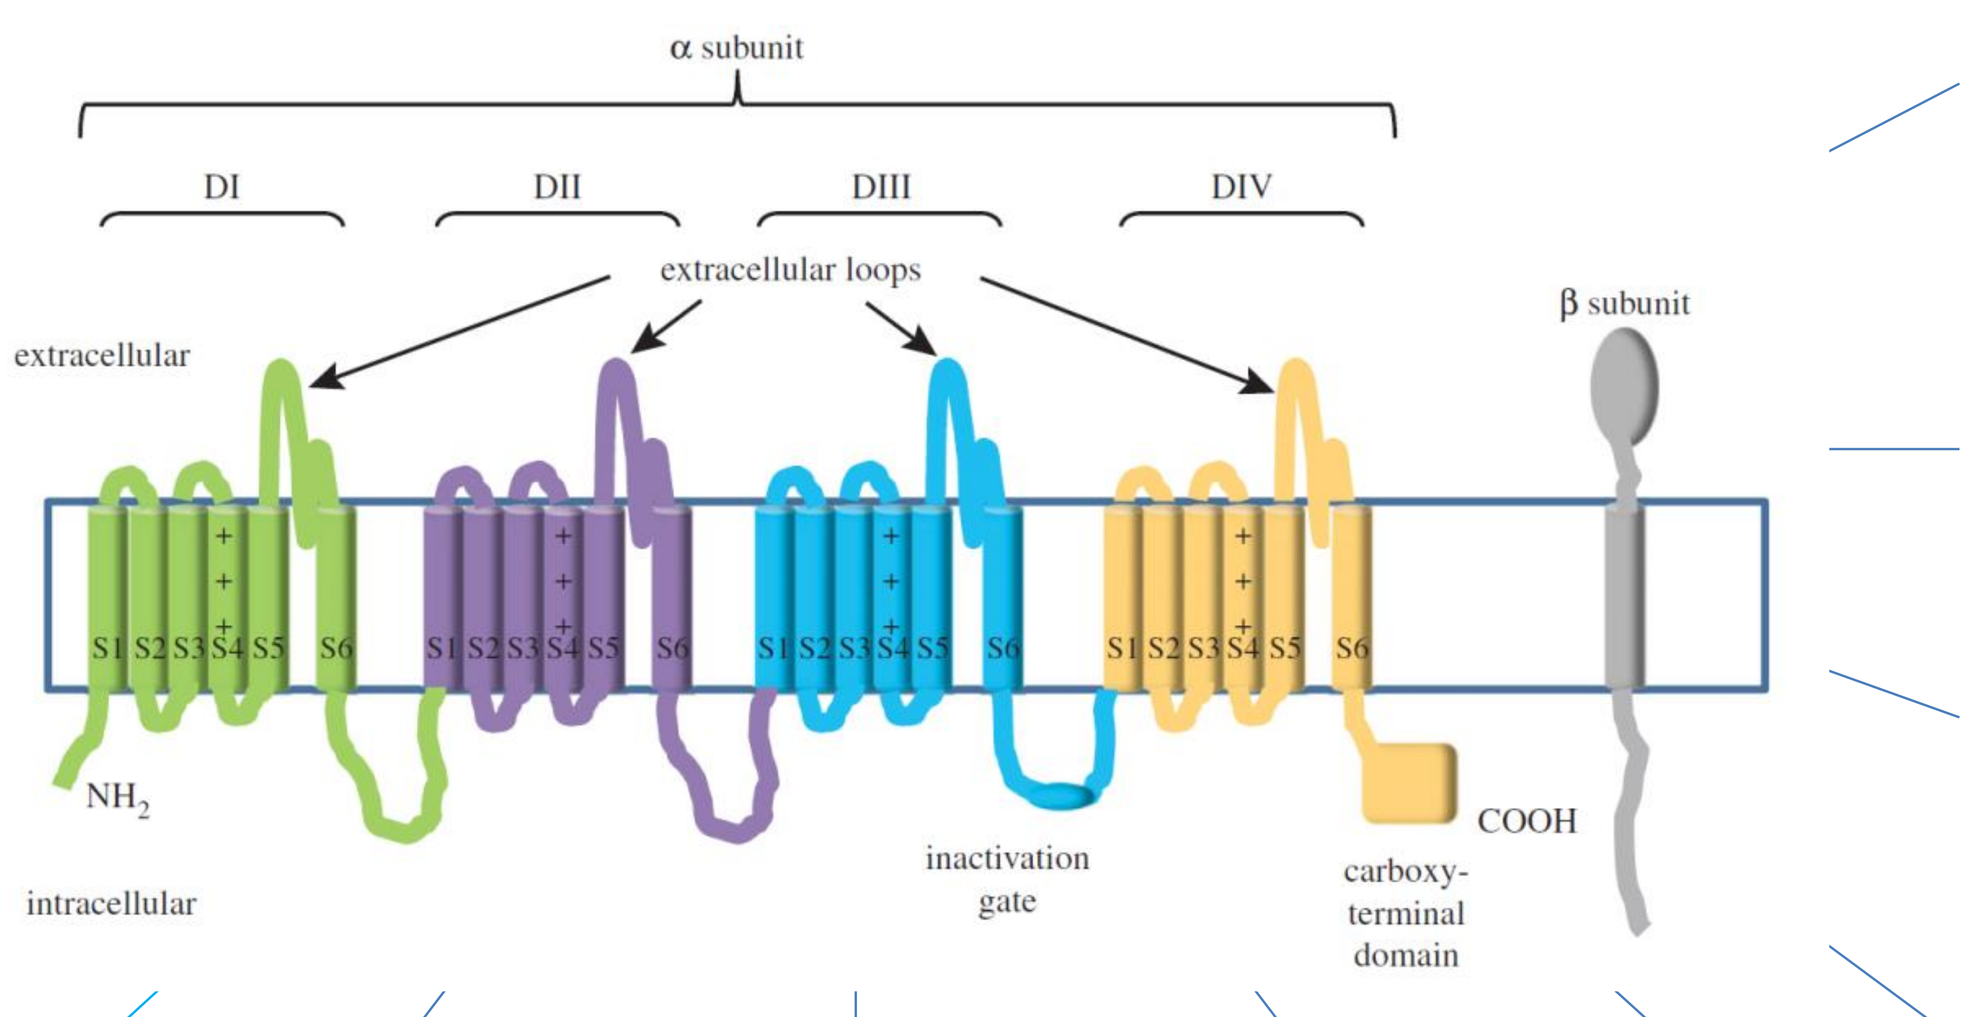
α subunit
DI
DII
DIII
DIV
extracellular loops
β subunit
extracellular
S1 S2 S3 S4 S5 S6
NH2
intracellular
inactivation gate
COOH
carboxy-terminal domain

Spider venom provides an amount of highly specific inhibitory cystine knots (ICK)-patterned toxins inhibiting $Na_V1.7$ with a natural selectivity against $Na_V1.5$ (> 2 logs). The BDS-1 and the Ssm6a are provided from sea anemone and Chinese red-headed centipede venoms, respectively.

In-vitro selectivity characterization of CyrTx-1a

The peptide exhibits nanomolar range affinity for $Na_V1.7$, $Na_V1.1$-1.3 and $Na_V1.6$ subtypes and micromolar range affinity for $Na_V1.5$ and $Na_V1.4$ subtypes in HEK293-Na_V engineered cell lines.

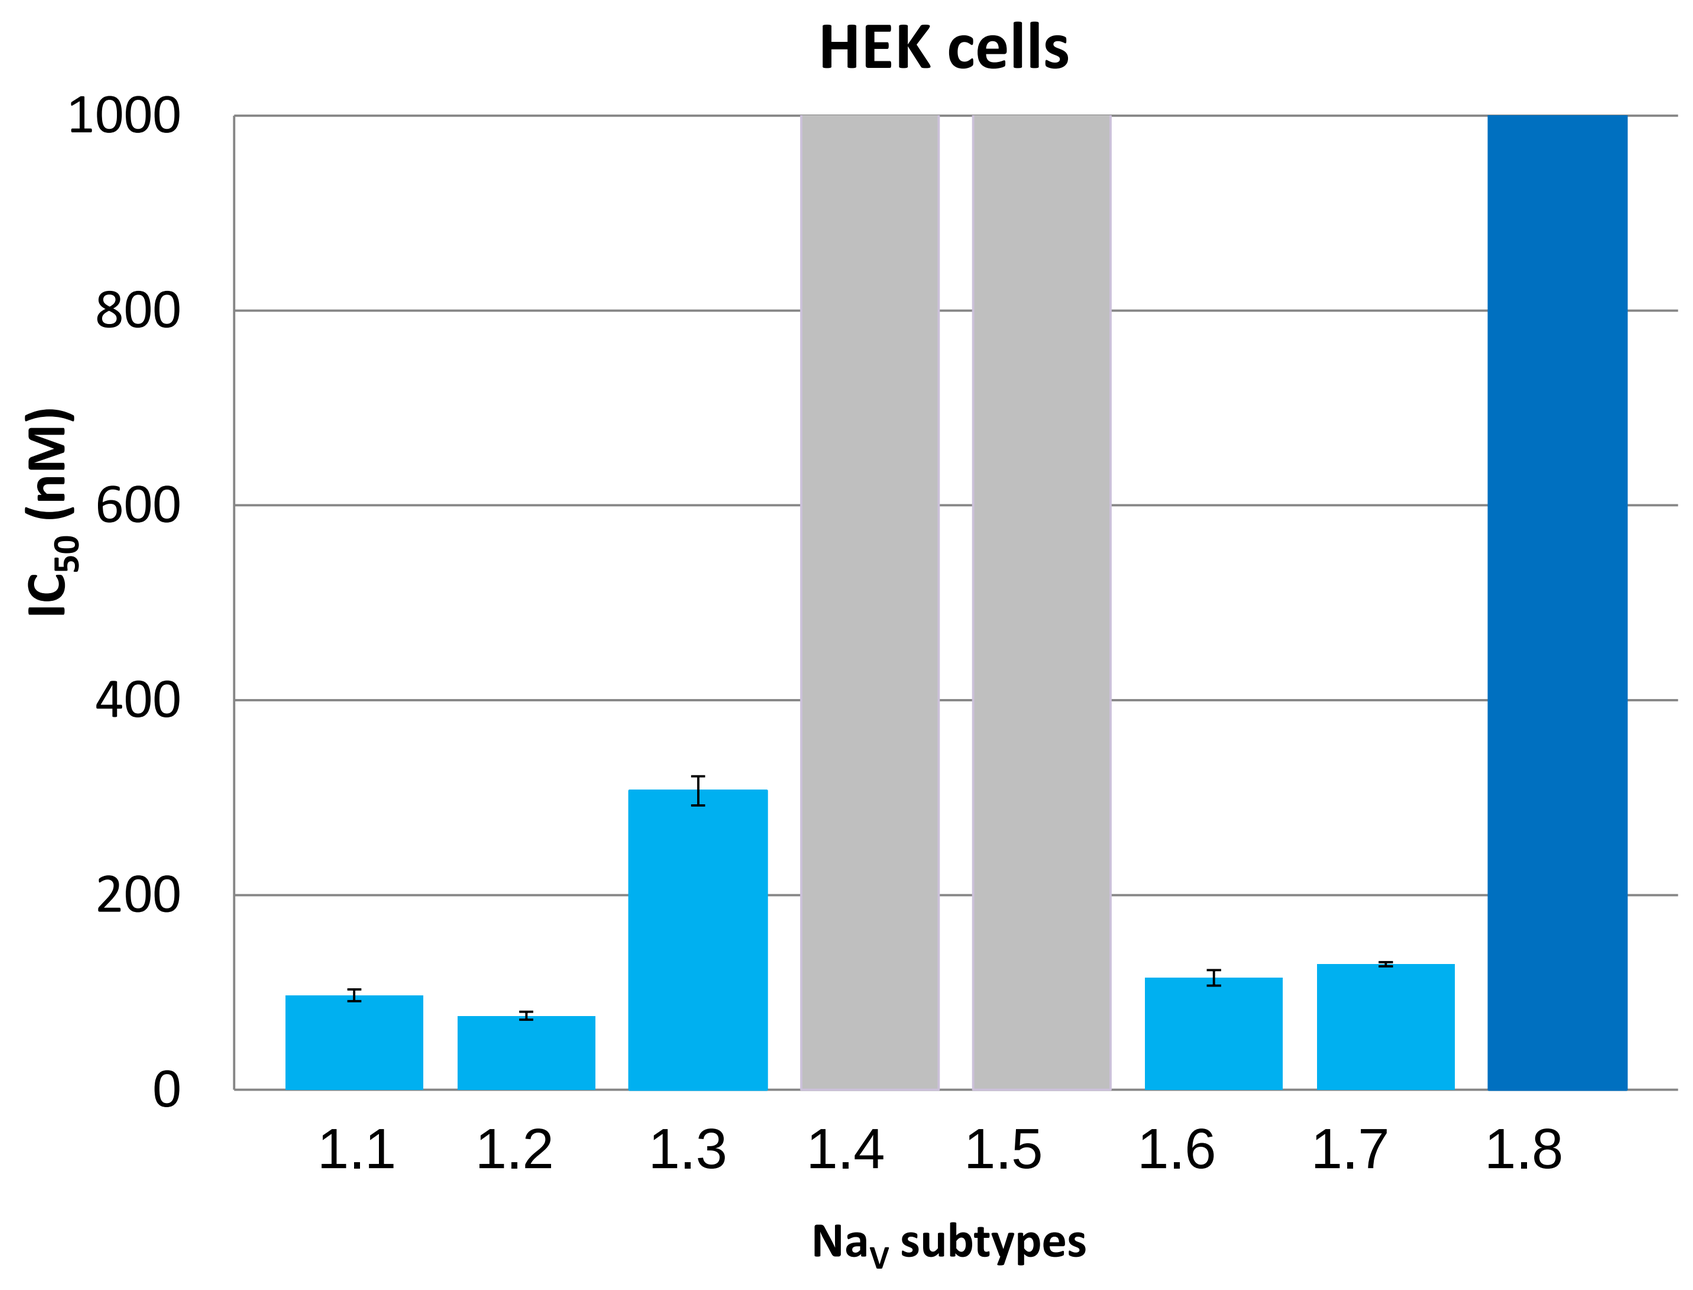
HEK cells
IC50 (nM)
0
200
400
600
800
1000
1.1
1.2
1.3
1.4
1.5
1.6
1.7
1.8
NaV subtypes

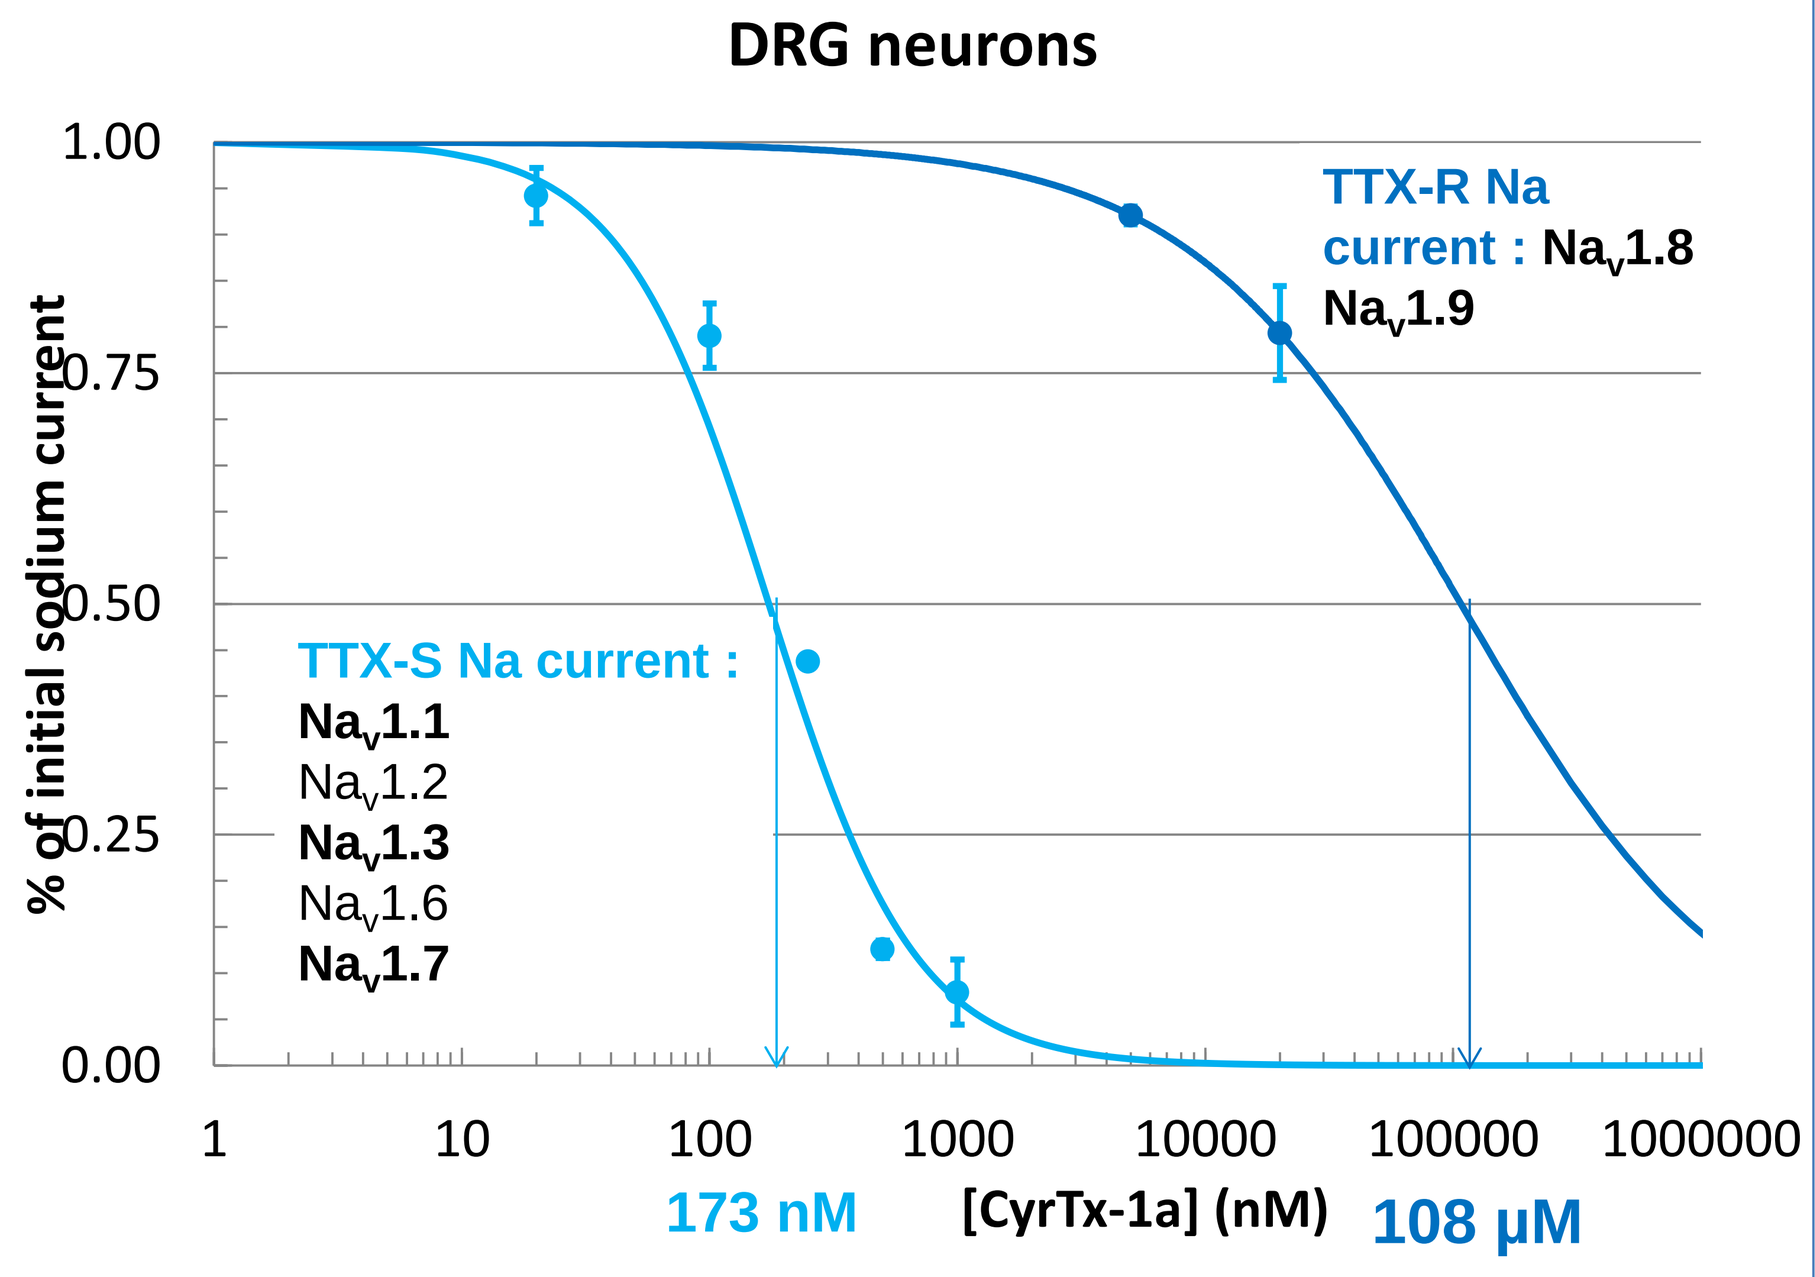
DRG neurons
% of initial sodium current
1.00
0.75
0.50
0.25
0.00
TTX-R Na current : NaV1.8
NaV1.9
TTX-S Na current :
NaV1.1
NaV1.2
NaV1.3
NaV1.6
NaV1.7
1
10
100
1000
10000
100000
1000000
173 nM
[CyrTx-1a] (nM)
108 µM

In DRG neurons isolated from adult mice, the CyrTx-1a peptide preferentially inhibited with very high affinity tetrodotoxin (TTX)-sensitive Na_V currents, mainly represented by the $Na_V1.7$ subtype.

In-vivo mouse studies using CyrTx-1a

- **Evaluation of painkiller effects**

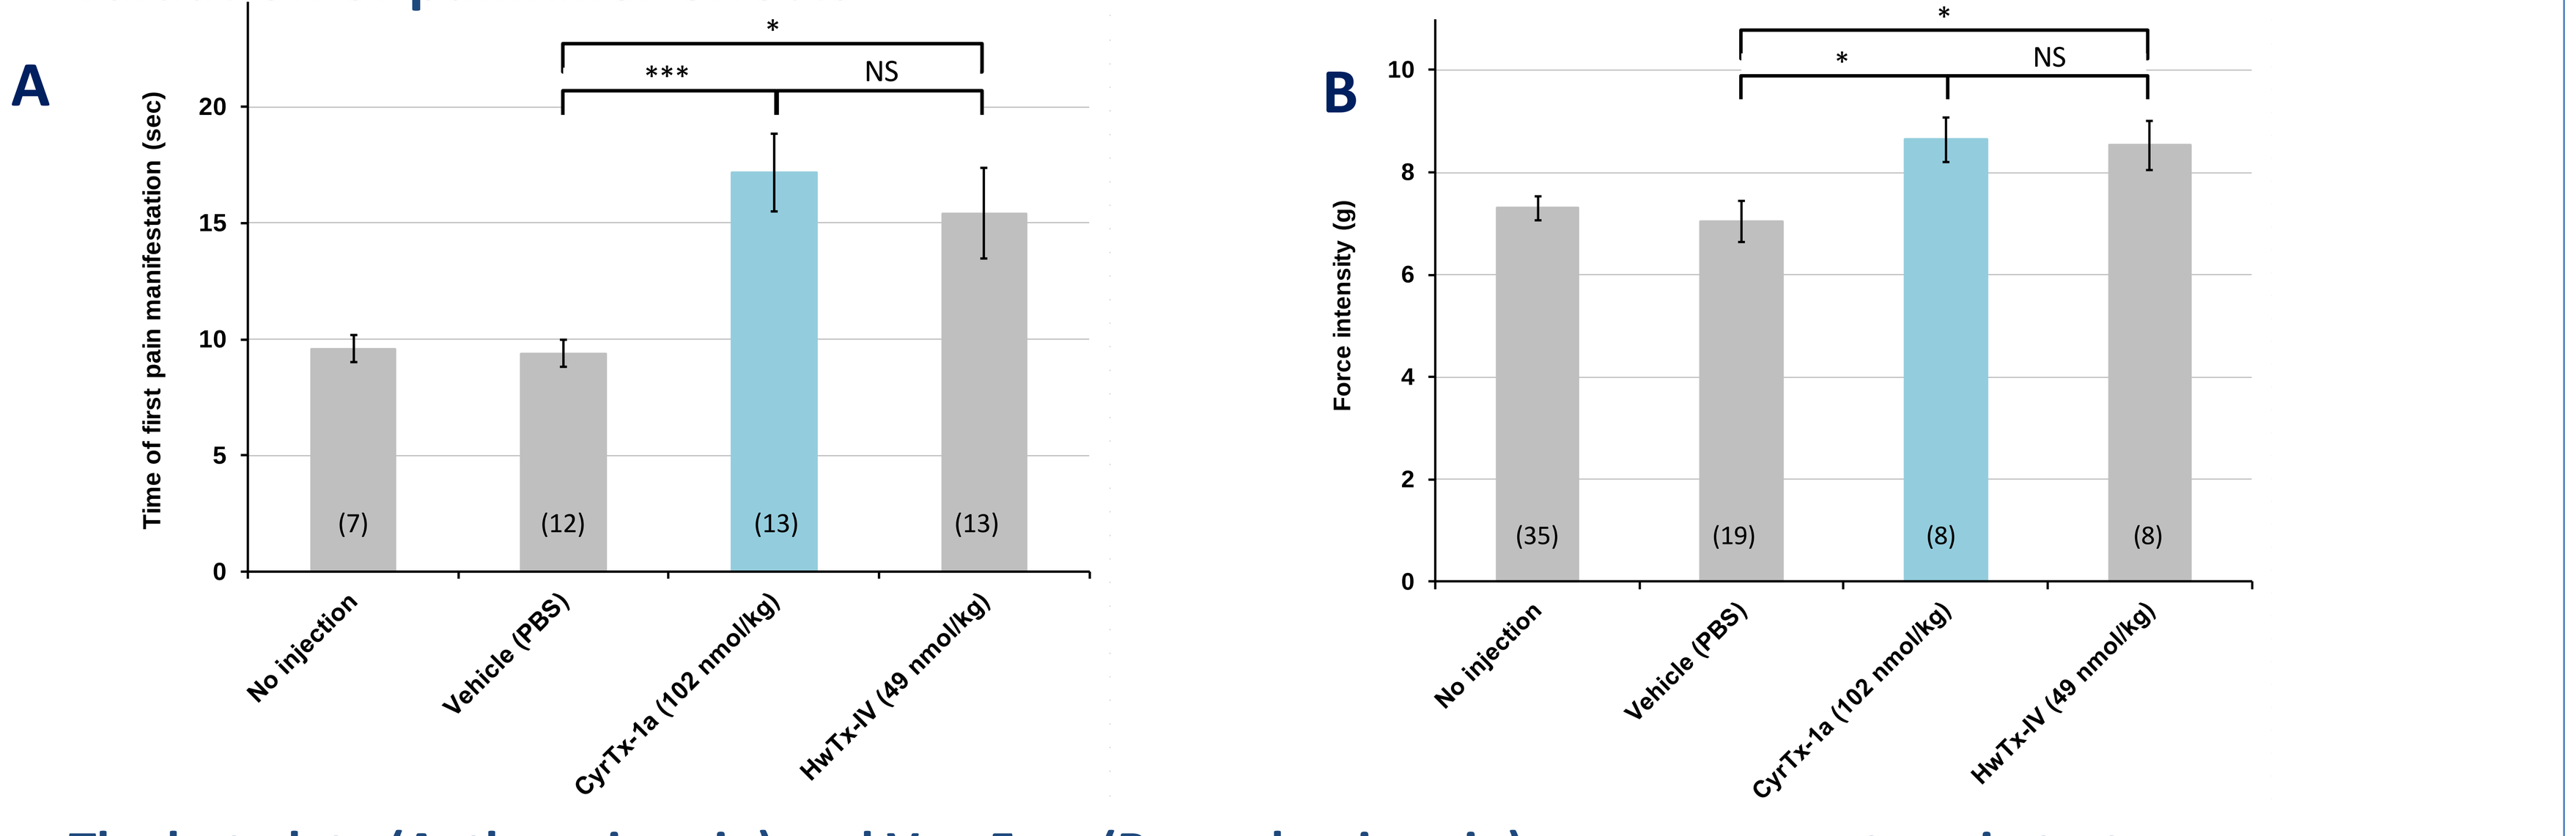
A
Time of first pain manifestation (sec)

NS
*
(7)
(12)
(13)
(13)
No injection
Vehicle (PBS)
CyrTx-1a (102 nmol/kg)
HwTx-IV (49 nmol/kg)
B
Force intensity (g)
*
NS
*
(35)
(19)
(8)
(8)

The hot plate (A, thermic pain) and Von Frey (B, mechanic pain) assays are acute pain tests.
The hot plate and von Frey pain assays showed that mice injected with 367 µg/kg of CyrTx-1a or with 200 µg/kg of HwTx-IV were more resistant to pain than animals injected with the vehicle (PBS).

- **Evaluation of muscle side-effects**

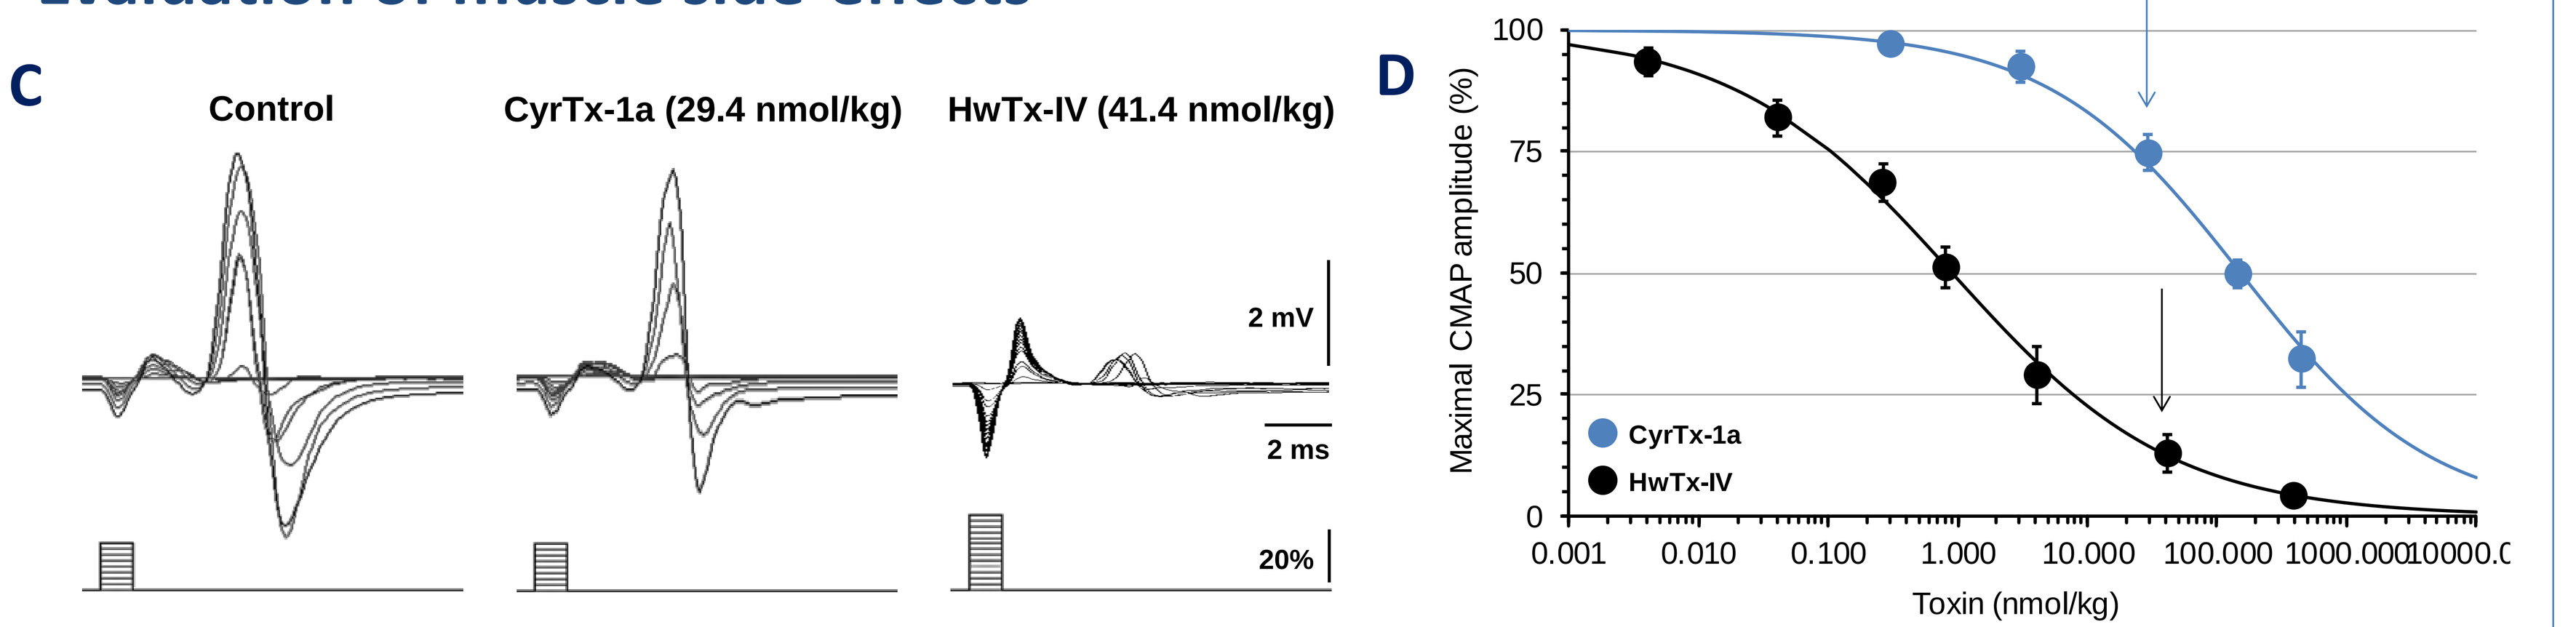
C
Control
CyrTx-1a (29.4 nmol/kg)
HwTx-IV (41.4 nmol/kg)
2 mV
2 ms
20%
D
Maximal CMAP amplitude (%)
CyrTx-1a
HwTx-IV
Toxin (nmol/kg)

The compound muscle action potential (C) assessment of both toxin reveals a narrow therapeutic window (D) with lethality occurring after the painkiller dose probably due to muscles (including diaphragm) paralysis.

Take home messages

- **As $Na_V1.7$ is involved in a multitude of genetic pain disorders, it seems obvious that this sodium channel subtype might be an important antinociceptive target.**
- **Toxins from spider venoms are valuable tools with characteristics of high affinity against $Na_V1.7$ and high selectivity against $Na_V1.5$. The studies of all these toxin sequences will help us better understand which amino acids are crucial for the binding on $Na_V1.7$ and which could increase selectivity against closely-related ion channels.**
- **CyrTx-1a is an promising toxin targeting $Na_V1.7$ with a nanomolar potency (selective vs. TTX-R NaV channels) and exhibiting antinociceptive *in-vivo* effects. However some structure-activity relationship studies are required to exclude the muscle contraction side-effects probably caused by the high affinity of CyrTx-1a for $Na_V1.6$ expressed in nodes of Ranvier of alpha motoneurons.**